Long-Acting Anti-Matriptase-2 Antibody As a Potentially Best-in-Class Therapy for Iron Overload Diseases

Authors, M. MA[1], M. HASKELL[1], D. SHERIDAN[1], S. ARROL[1], Z. CHENG[1], and A. HAYWARD[1]

1. Rallybio IPE LLC, New Haven, Connecticut, USA

INTRODUCTION AND OBJECTIVE

Iron overload can lead to severe and life-threatening conditions. Many diseases and conditions are associated with iron overload, including polycythemia vera (PV), myelodysplastic syndrome (MDS) and beta-thalassemia (BT).

The current standard of care for iron overload diseases primarily revolves around reducing the excess iron accumulated in the body to prevent organ damage. These treatments carry risks like anemia and present some side effects.

Therapeutic Mechanism:
Hepcidin, a key regulatory hormone in iron metabolism, is critical in maintaining iron homeostasis within the body.
Matriptase 2 (MTP-2) is a serine protease that plays an essential role in hepcidin formation.
Inhibition of MTP-2 to increase levels of hepcidin.

RLYB332 is a fully-humanized MAb inhibitor of MTP-2. The mechanism of action supports the development for the treatment of iron overload and ineffective erythropoiesis-associated disorders such as PV, MSD, or BT.

Objective:
To evaluate the pharmacokinetic (PK), pharmacodynamic (PD), and safety profiles of the anti-MTP-2 antibody RLYB332 using a humanized FcRn (Tg32) mouse model.

METHOD

Model

A humanized FcRn(Tg32) mouse was used to approximate human physiological responses.

Administration

RYB332, R79, and M03 were administered via intravenous injection.

Group settings:

- RLYB332: at dosages of 1 and 3 mg/kg/day;
- Comparable molecules: R79 (1 and 3 mg/kg/day) and M03 (1 and 3 mg/kg/day).

Table 1. The study design of PK and PD in Tg 32 mouse model

Group No.	Test Material	Dose Level (mg/kg/day)	Dose Volume (mL/kg)	No. of Animals PK	No. of Animals PD
1	Control	0	5	15	9
2	RLYB332	1	5	15	9
3	RLYB332	3	5	15	9
4	R79	1	5	15	9
5	R79	3	5	15	9
6	M03	1	5	15	9
7	M03	3	5	15	9

RESULTS

- **Safety:**
 - There was no unscheduled mortality and no test item-related clinical observations.
 - There were no test item-related effects on body weights.
 - Following macroscopic examination of gross lesions, no test item-related effects were found.

- **PK:**

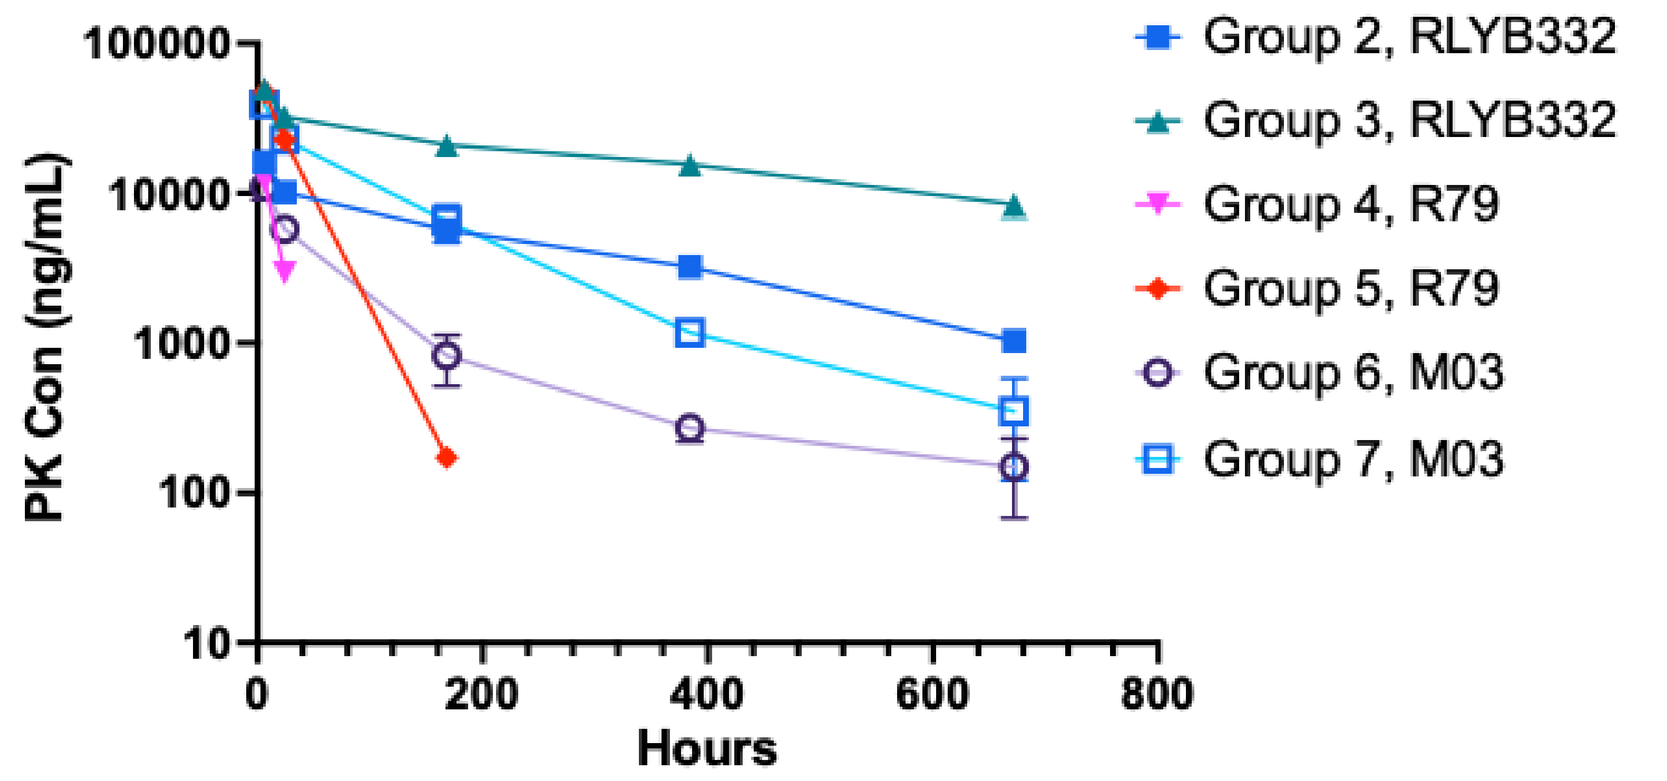
PK Con (ng/mL)
100000
10000
1000
100
10
0
200
400
600
800
Hours
Group 2, RLYB332
Group 3, RLYB332
Group 4, R79
Group 5, R79
Group 6, M03
Group 7, M03

Figure 1. PK profiles of RYB332, R79, and M03 in Tg32 mice following intravenous administration at 1 and 3 mg/kg/day (N=3).

Table 2. The PK parameters of RYB332, R79, and M03 in Tg32 mice following intravenous administration at 1 and 3 mg/kg/day.

Analytes	Group	Number of Time-points	Dose	Rsq	$T_{1/2}$	T_{max}	C_{max}
			mg/kg/day			hour	ng/mL
RLYB332	2	5	1	0.99	204	6	16189.1
RLYB332	3	5	3	0.99	348	6	49598.1
RG79	4	2	1	NC	NC	6	12038.6
RG79	5	3	3	1.00	20	6	47330.6
M03	6	5	1	0.94	209	6	11036.2
M03	7	5	3	0.97	121	6	39057.2

- **PD:**

Figure 2. Percentage change of serum iron over time

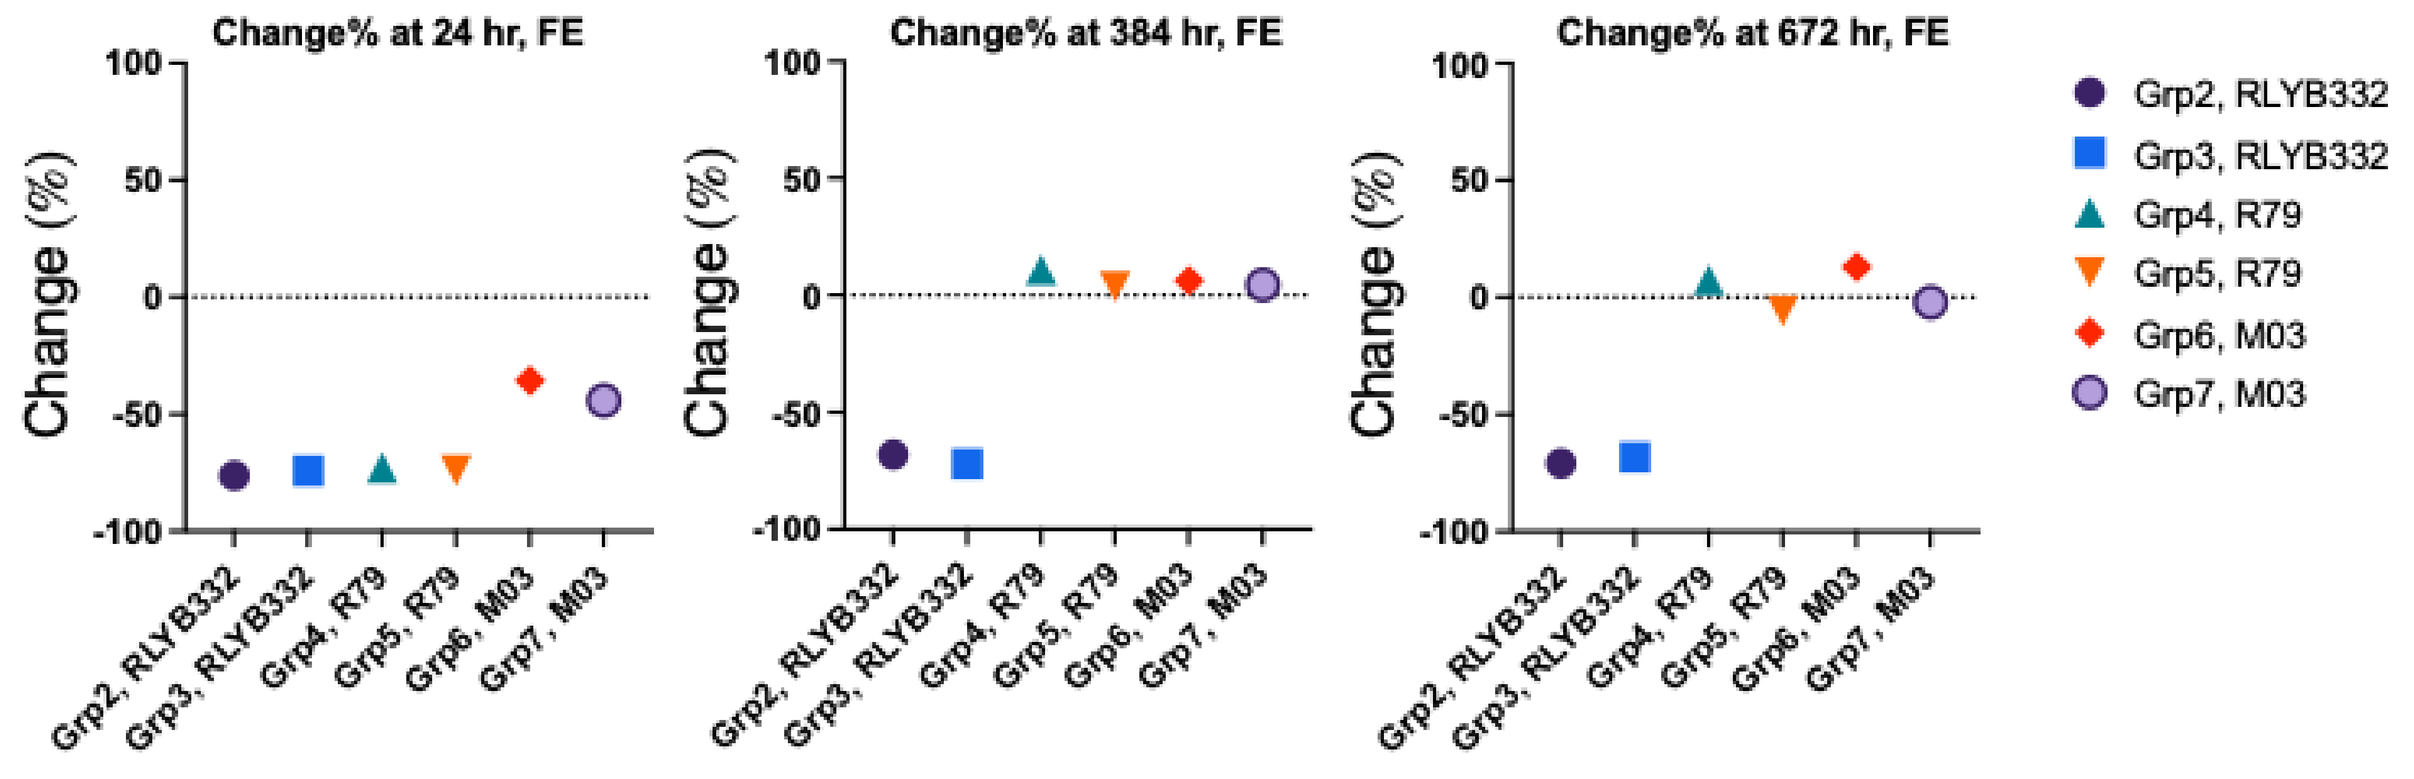
Change% at 24 hr, FE
Change% at 384 hr, FE
Change% at 672 hr, FE
Change (%)
Grp2, RLYB332
Grp3, RLYB332
Grp4, R79
Grp5, R79
Grp6, M03
Grp7, M03

Figure 3. Percentage change of UIBC over time

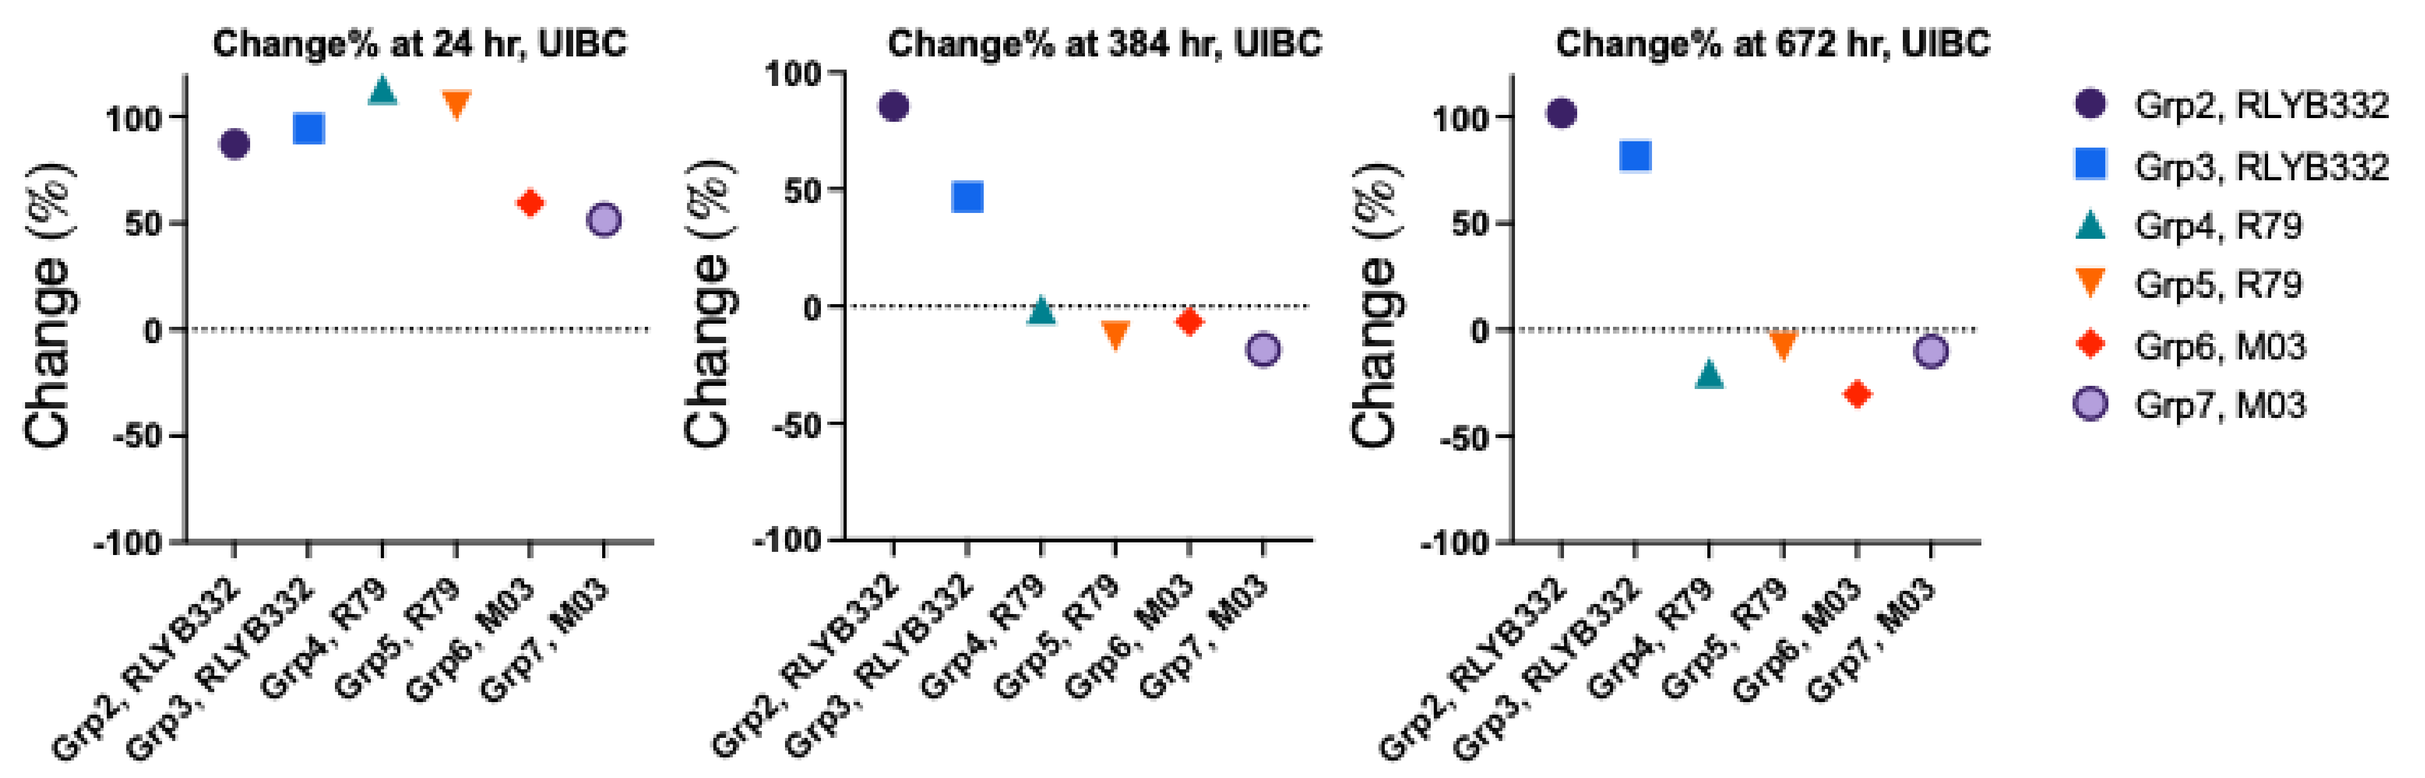
Change% at 24 hr, UIBC
Change% at 384 hr, UIBC
Change% at 672 hr, UIBC
Change (%)
Grp2, RLYB332
Grp3, RLYB332
Grp4, R79
Grp5, R79
Grp6, M03
Grp7, M03

Figure 4. Percentage change of TSAT over time

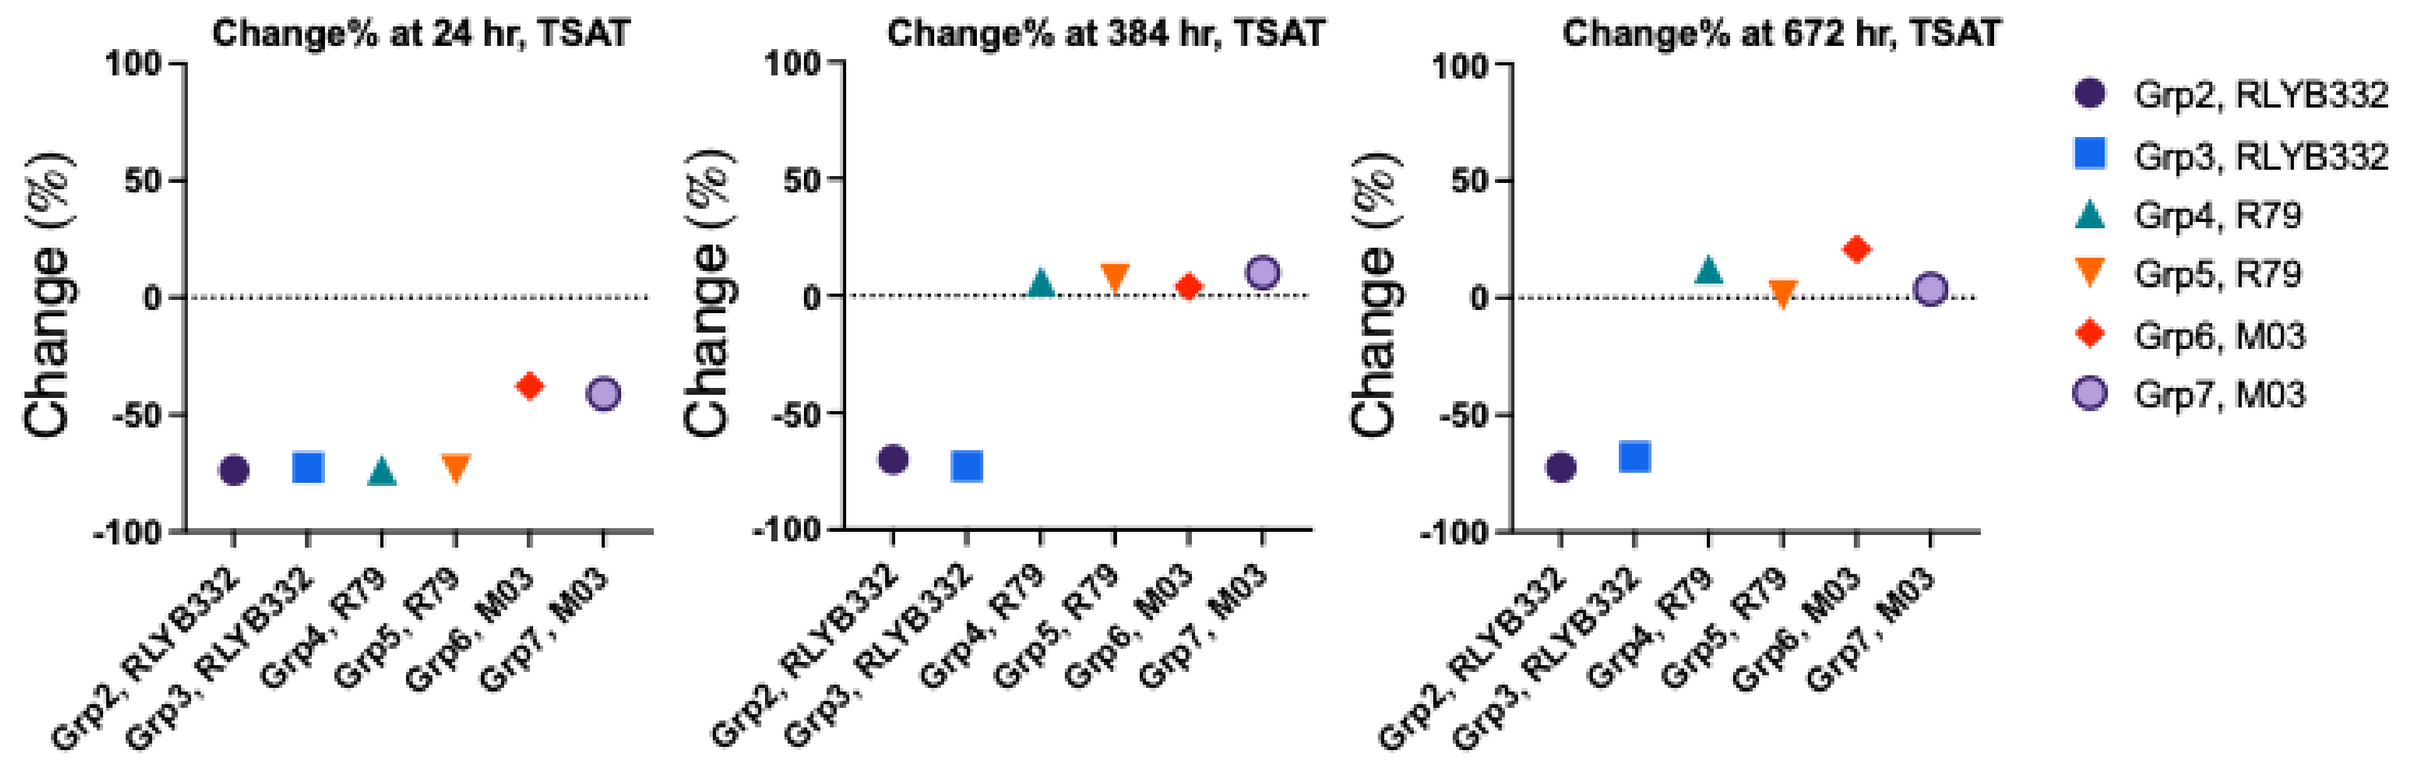
Change% at 24 hr, TSAT
Change% at 384 hr, TSAT
Change% at 672 hr, TSAT
Change (%)
Grp2, RLYB332
Grp3, RLYB332
Grp4, R79
Grp5, R79
Grp6, M03
Grp7, M03

Table 3. Summary of PD results observed on serum iron, UIBC, and TSAT levels.

	Resutls (Mean)			% Change from the baseline		
	24 hr	384 hr	672 hr	24 hr	384 hr	672 hr
Control						
FE (µg/dL)	173.0	154.7	159.0	NC	NC	NC
UIBC (µg/dL)	115.0	139.3	129.3	NC	NC	NC
TSAT (%)	60.7	52.9	55.1	NC	NC	NC
RLYB332 (1mg/kg)						
FE (µg/dL)	41.0	49.3	46.3	-76.3	-68.1	-70.9
UIBC (µg/dL)	215.3	258.3	261.0	87.2	85.4	101.8
TSAT (%)	16.0	15.9	15.2	-73.7	-69.9	-72.4
RLYB332 (3mg/kg)						
FE (µg/dL)	44.7	43.3	50.3	-74.2	-72.0	-68.3
UIBC (µg/dL)	223.7	260.3	235.0	94.5	46.8	81.7
TSAT (%)	16.7	14.4	17.7	-72.5	-72.8	-67.9
R79 (1mg/gk)						
FE (µg/dL)	47.0	171.7	171.0	-72.8	11.0	7.5
UIBC (µg/dL)	245.3	136.3	103.3	113.3	-1.2	-20.1
TSAT (%)	16.1	56.2	62.0	-73.5	6.3	12.4
R79 (3mg/kg)						
FE (µg/dL)	45.0	160.3	149.7	-74.0	3.7	-5.9
UIBC (µg/dL)	235.7	121.7	118.7	104.9	-13.0	-8.2
TSAT (%)	16.0	56.7	55.7	-73.6	7.1	1.1
M03 (1mg/kg)						
FE (µg/dL)	111.3	164.3	179.7	-35.6	6.3	13.0
UIBC (µg/dL)	183.3	131.3	90.3	59.4	-6.6	-30.2
TSAT (%)	37.8	55.0	66.6	-37.7	3.9	20.8
M03 (3mg/kg)						
FE (µg/dL)	96.7	161.7	155.7	-44.1	4.5	-2.1
UIBC (µg/dL)	174.3	115.0	116.3	51.6	-18.5	-10.1
TSAT (%)	35.7	58.1	57.2	-41.1	9.8	3.8

CONCLUSIONS

RLYB332, the antibody targeting MTP-2, is a promising candidate for treatment of iron overload disorders.

In our study, single intravenous injections of the antibody were well tolerated in humanized FcRn mice at doses tested, whilst demonstrating greater effects on PD parameters than compared molecules.

Rapid and sustained pharmacodynamic effects were observed in both the 1 mg/kg and 3 mg/kg groups, highlighting the potential for self-administration in patients with iron overload disorders.

These findings support the continued preclinical and clinical development of this **anti-MTP-2 antibody as a potentially best-in-class therapeutic molecule** for managing iron overload diseases**.**

REFERENCES

1. Valenti, L., Rametta, R., Dongiovanni, P., Motta, B. M., Canavesi, E., Pelusi, S., Pulixi, E. A., Fracanzani, A. L. & Fargion, S. The A736V TMPRSS6 polymorphism influences hepatic iron overload in nonalcoholic fatty liver disease. PLoS One 7, e48804 (2012).
2. Tanaka, T., Roy, C. N., Yao, W., Matteini, A., Semba, R. D., Arking, D., Walston, J. D., Fried, L. P., Singleton, A., Guralnik, J., Abecasis, G. R., Bandinelli, S., Longo, D. L. & Ferrucci, L. A genome-wide association analysis of serum iron concentrations. Blood 115, 94-96 (2010).
3. Ganz, T., Nemeth, E., Rivella, S., Goldberg, P., Dibble, A. R., McCaleb, M. L., Guo, S., Monia, B. P. & Barrett, T. D. TMPRSS6 as a Therapeutic Target for Disorders of Erythropoiesis and Iron Homeostasis. Adv Ther 40, 1317-1333 (2023).
4. Nai, A., Pagani, A., Mandelli, G., Lidonnici, M. R., Silvestri, L., Ferrari, G. & Camaschella, C. Deletion of TMPRSS6 attenuates the phenotype in a mouse model of beta-thalassemia. Blood 119, 5021-5029 (2012).

ACKNOWLEDGEMENTS

Charles River Laboratories, Sherbrooke, QC, Canada

CONTACT INFORMATION

Email: mma@rallybio.com; Tel: 1-805-410-2415